Validation of Assessments for Reward Processing for Use in a Phase 2 Study of a Novel Histamine H3 Inverse Agonist, ALTO-203, in Major Depression

Nicholas J. Cooper[1], Josh T. Jordan[1], Quentin J. M. Huys[2], Greg Hajcak[3], Mike Avissar[1], Jessica Powell[1], Amit Etkin[1], Adam J. Savitz[1].
[1]Alto Neuroscience, Inc., Mountain View, CA | [2]University College London, Psychiatry, London, United Kingdom | [3]Santa Clara University, School of Education and Counseling Psychology, Santa Clara, United States

1. Background

- Anhedonia, the lack of pleasure or motivation, may reflect dopaminergic dysfunction.
- The Histamine H3 receptor (H3R) is a brain-specific negative regulator of multiple neurotransmitter systems, including dopamine.
- Established tests of reward processing (e.g., PRT [1] and EEfRT [2]) tend to be lengthy and poorly tolerated. They may also have low test-retest reliability and inconsistent correlations with clinical constructs like anhedonia.

2. ALTO-203 Proof of Concept Study

- ALTO-203 is a novel H3R inverse agonist shown to increase dopamine release in the nucleus accumbens in rodents, and thus may improve reward sensitivity, motivation and anhedonia in MDD patients.
- To more effectively characterize pharmacodynamic changes in the POC study, we developed and validated two computerized tests for different aspects of reward processing and motivation and examined their relationship to anhedonia.

3. Development of Reward Tasks

- Effort Tapping (Figure 2) is a free-operant paradigm where participants can earn +10 (low reward) or +100 points (high reward) each time they press a key during periods of 3 to 17 seconds. Akin to findings with apathy [3], we hypothesized that participants with higher anhedonia would show a lower rate of pressing during the low reward (+10) versus high reward condition, indicating greater sensitivity to reward.

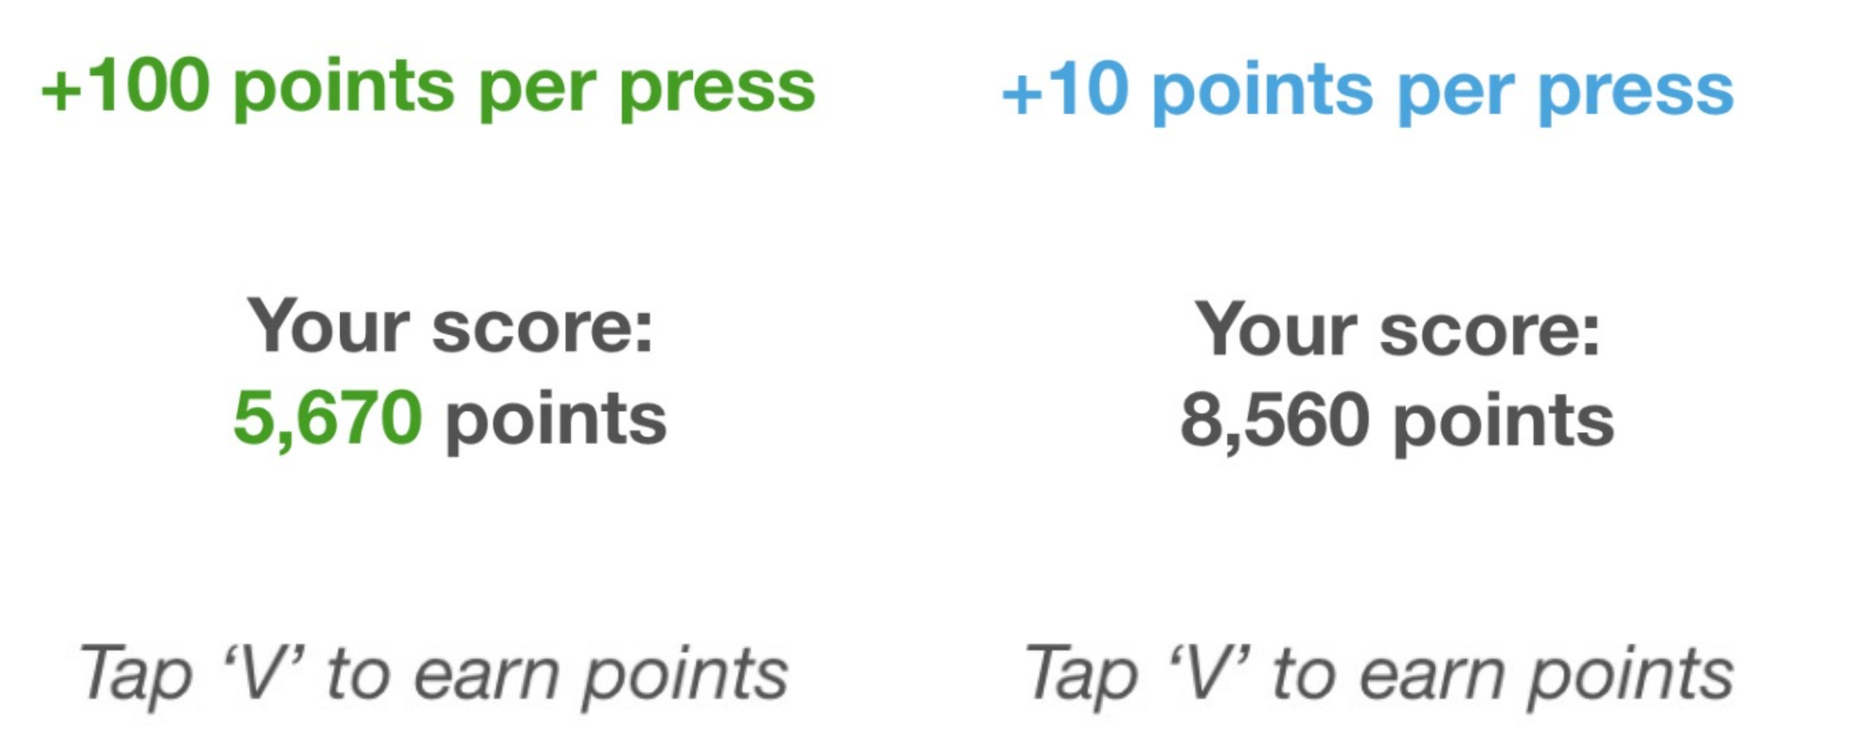
+100 points per press
+10 points per press
Your score:
5,670 points
Your score:
8,560 points
Tap ‘V’ to earn points
Tap ‘V’ to earn points

Figure 1. Screenshots for the Effort Tapping task. The left screen shows a trial in the high reward condition and the right screen shows the low reward condition. In each of 3 rounds participants aim to get 20,000 points.

- The Probabilistic Instrumental Learning Task (**PILT**, Figure 3) is an adaptation of a two-armed bandit test that measures the ability to learn arbitrary stimulus-reward probabilistic associations. The task involves multiple rounds with novel stimulus pairs becoming progressively harder to distinguish.
- We hypothesized that anhedonia would be associated with a lower learning rate (α) and increased decision noise (β) [4].

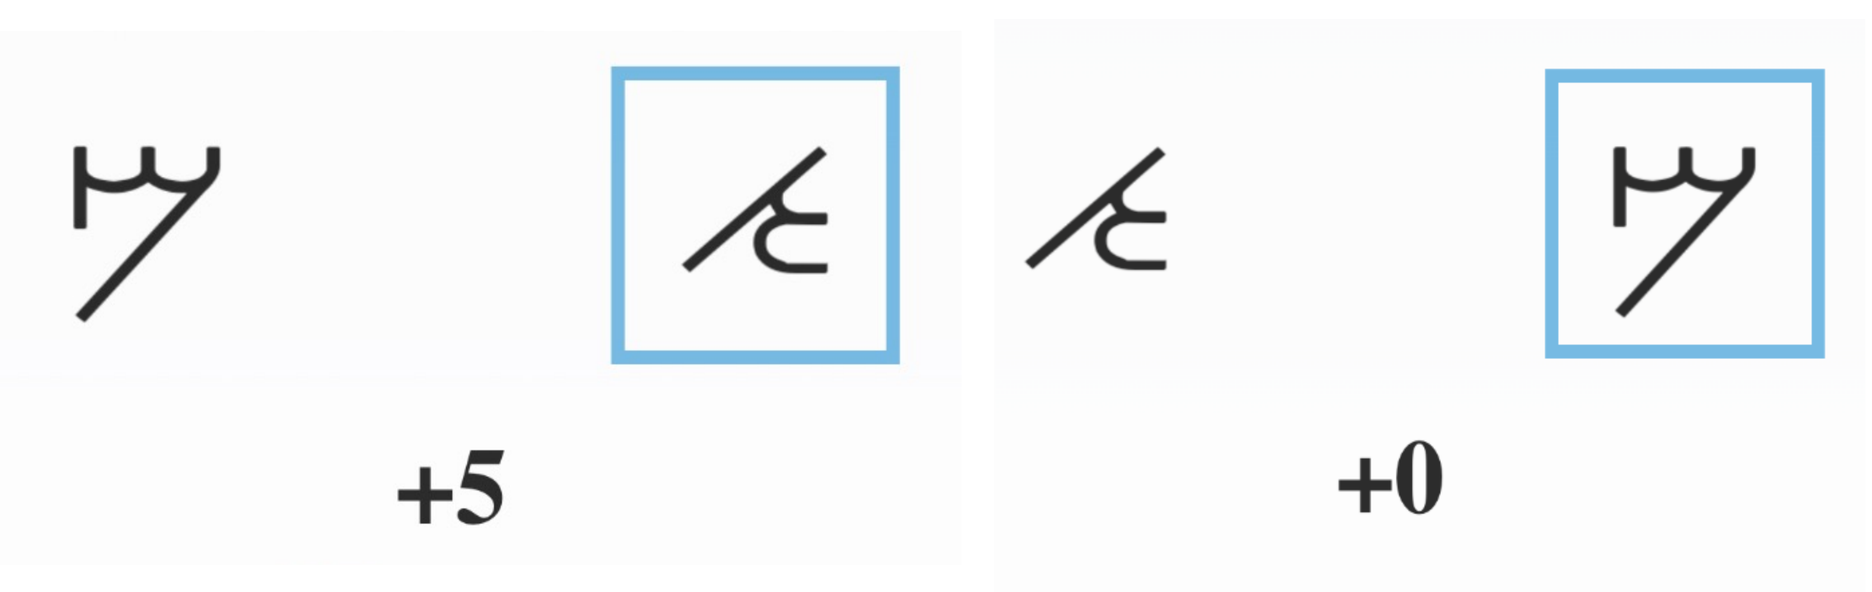
+5
+0

Figure 2. Screenshots for the PILT task. The left screen shows the ‘reinforced’ symbol being selected. The right screen shows a trial where the ‘non-reinforced’ symbol is selected. Left/right side presentation is randomized on each trial.

4. Test-retest reliability study

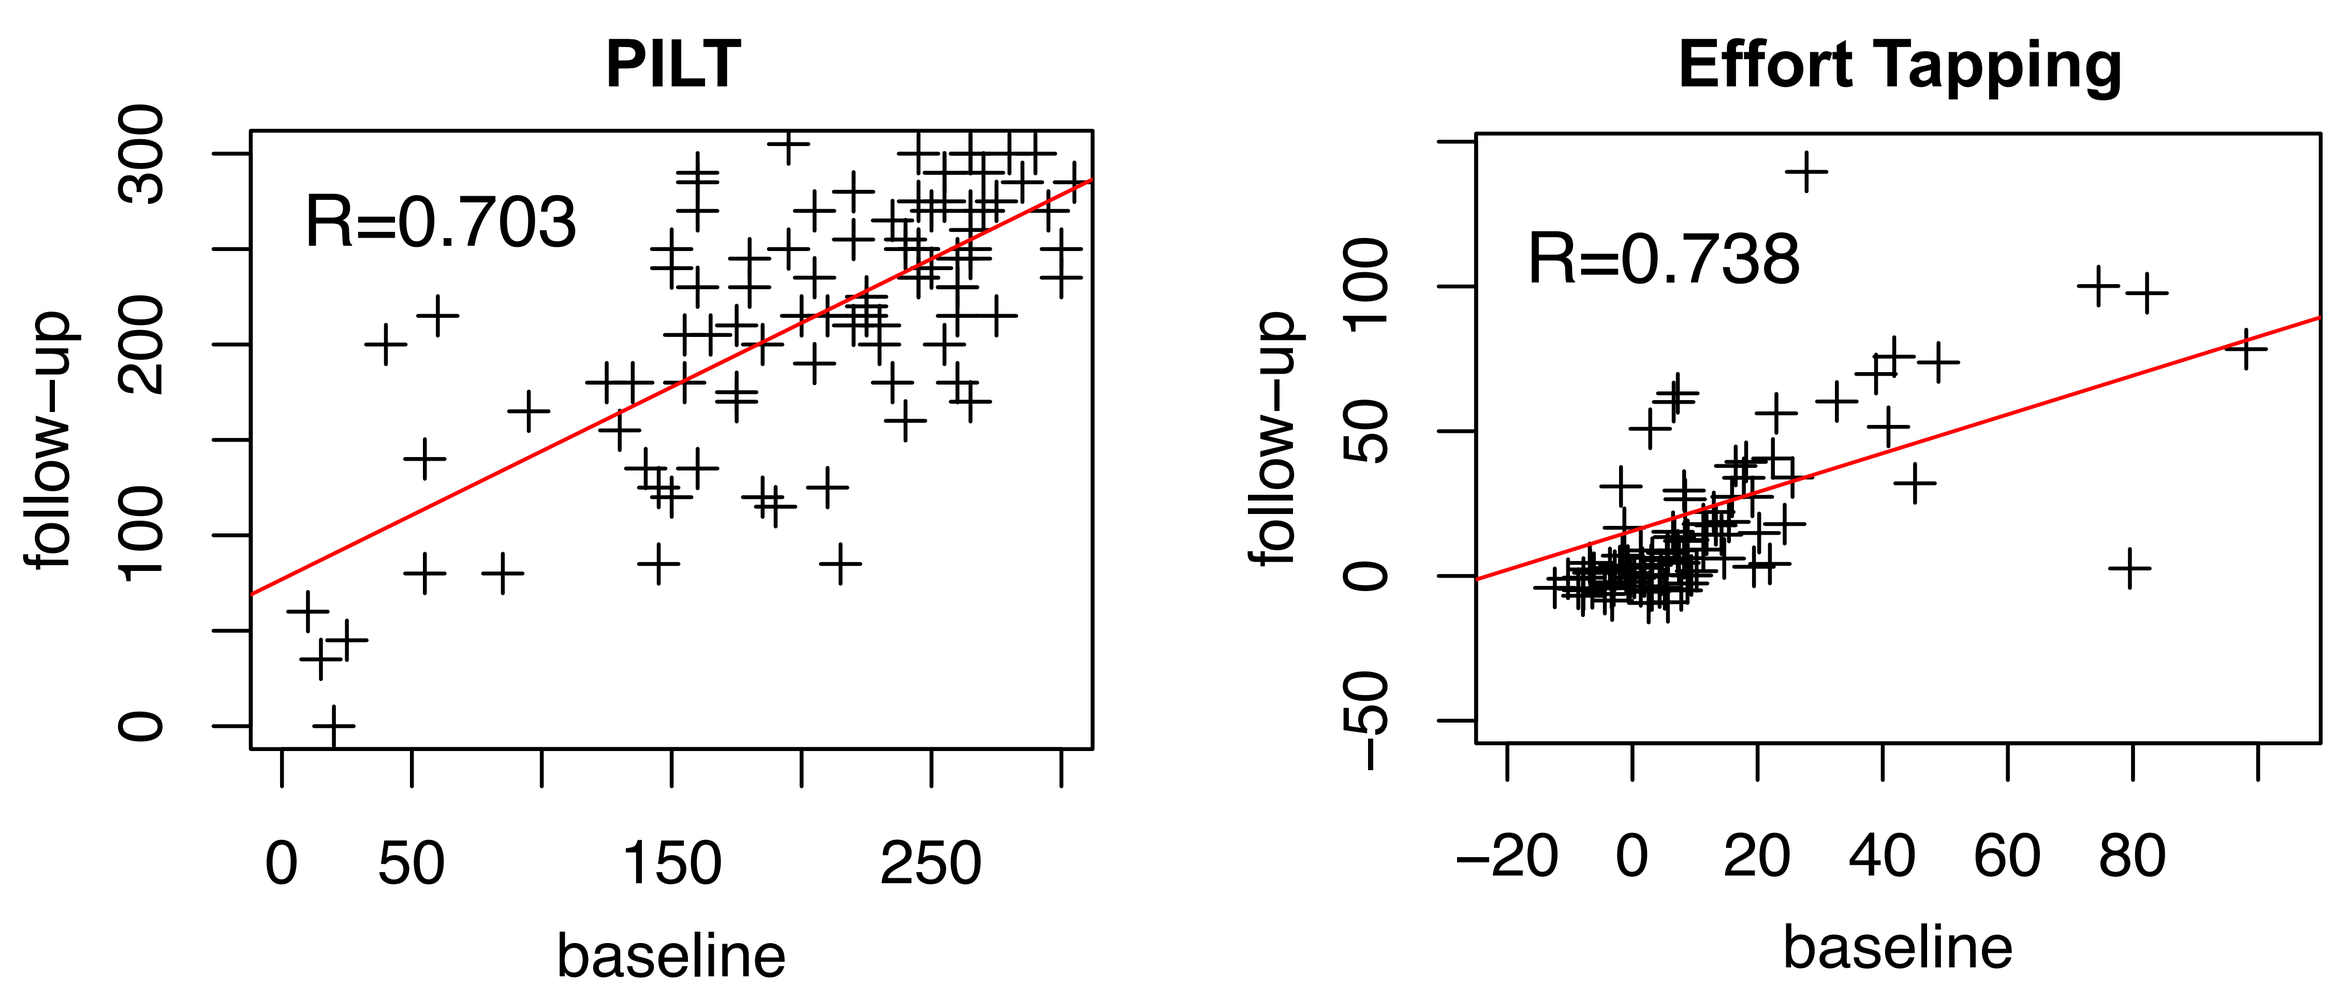
PILT
R=0.703
follow–up
baseline
Effort Tapping
R=0.738
follow–up
baseline

Figure 3. Scatterplots for test and retest on PILT decision noise and Effort Tapping latency difference with a two week delay in 73 subjects demonstrate that **these tasks, while shorter than the established tasks, are highly reliable.**

4. Construct and External Validation Studies

- After establishing strong test-retest reliability (ICC > 0.7), the new tasks, plus tasks and surveys from Table 1, were obtained for online samples to determine construct and external validity:
 - 168 participants, 18 - 70 years (mean 37.8), 60.1% female.
 - 378 participants, 18 - 73 years (mean 38.3), 56.9% female.
- The first study used real money to incentivize trials for EEfRT and PRT tasks, while the second only gave ‘points.’

Label	Test Name	Outcome variable
PRT	Probabilistic Reward Task [1]	Response bias ('Log b') towards the (long or short) mouth with higher reward probability across 2 rounds of 100 trials.
EEfRT	Effort Expenditure for Rewards Task [2]	Percentage of 50 administered trials choosing the hard (100 pinky taps) over the easy (30 taps) condition ('EEfRT % hard').
DDT	Delayed Discounting Task	ED50, delay duration at which $10 reward is considered of equal value to $5 now.
Tapping	Finger Tapping (sensorimotor)	Mean latency for 150 dominant hand taps.
Buckets	Filling buckets (patch foraging task)	Reward versus time spent; Time spent in patches
PHQ-8	Patient Health Questionnaire	Minus suicidality question from PHQ-9.
SHAPS	Snaith-Hamilton Pleasure Scale	Anhedonia scores flipped to match the direction of DARS/PVSS.
DARS	Dimensional Anhedonia Rating Scale [5]	Lower scores indicate greater anhedonia.
PVSS	Positive Valence Systems Scale [6]	Lower scores indicate greater anhedonia.
BIS/BAS	Behavioral Inhibition/Activation System [7]	Measures sensitivity to reward (BAS) and punishment (BIS)
TEPS	Temporal Experience of Pleasure Scale [8]	Anticipatory and consummatory pleasure; higher scores indicate greater experience of pleasure.
PANAS	Positive and Negative Affect Scale [9]	Acute measure of affect ‘right now’, separate positive and negative subscales.

Table 1. Listing of comparison tasks and scales collected alongside the new reward tasks in the validation studies. TEPS and PANAS were only used in the second study.

5. Tolerability

- Participants rated tasks on a visual 5-point scale (pleasantness). **The new tasks were substantially more tolera5-point established tasks**, Effort Tapping (p < .05, d=0.15) versus EEfRT, and PILT versus PRT (p < .001, d=0.6).
- **Median time taken for the new tasks was substantially shorter than established tasks**: 3.6 minutes for Effort Tapping (vs. 20.7 minutes for EEfRT) and 4 minutes for PILT (vs.17.2 minutes PRT).

6. Relationship Between New and Established Tasks

- **PILT decision noise correlated with PRT logB** in both the first (r = 0.26, p < .01) and second (r = 0.18, p < .01) studies.
- No significant relationship with our reward tasks and EEfRT were observed, however there were correlations with DDT-ED50 (p < .05) and Finger Tapping (p < .001).
- PILT decision noise correlated with Effort Tapping latency difference (r = 0.22, p < .001) suggesting a conceptual overlap.

7. Relationship between Tasks and Anhedonia

- **Performance in the new reward tasks was significantly and reproducibly correlated with anhedonia, positive emotion, and BIS/BAS measures** [~0.15 - 0.3], in both studies. Correlations from the second study are presented in Figure 4.
- **PRT and EEfRT were inconsistently related to anhedonia-related constructs.** PRT or EEfRT were not correlated with anhedonia in the first study, when monetary incentives were employed [10]. In the second study there were weak associations between EEfRT and BIS/BAS (p < .05), PILT and TEPS ‘anticipatory’ scale (p < .05).
- In both studies, **Effort Tapping was the task most broadly associated with scales related to anhedonia** (DARS, PVSS, TEPS), excepting the SHAPS.
- **PILT correlated with BIS/BAS in both studies**.
- PILT and Effort Tapping correlated with PANAS positive affect.
- **No correlations were seen between Effort Tapping or PILT with depression/anxiety symptoms on the PHQ-8 or GAD-7**.

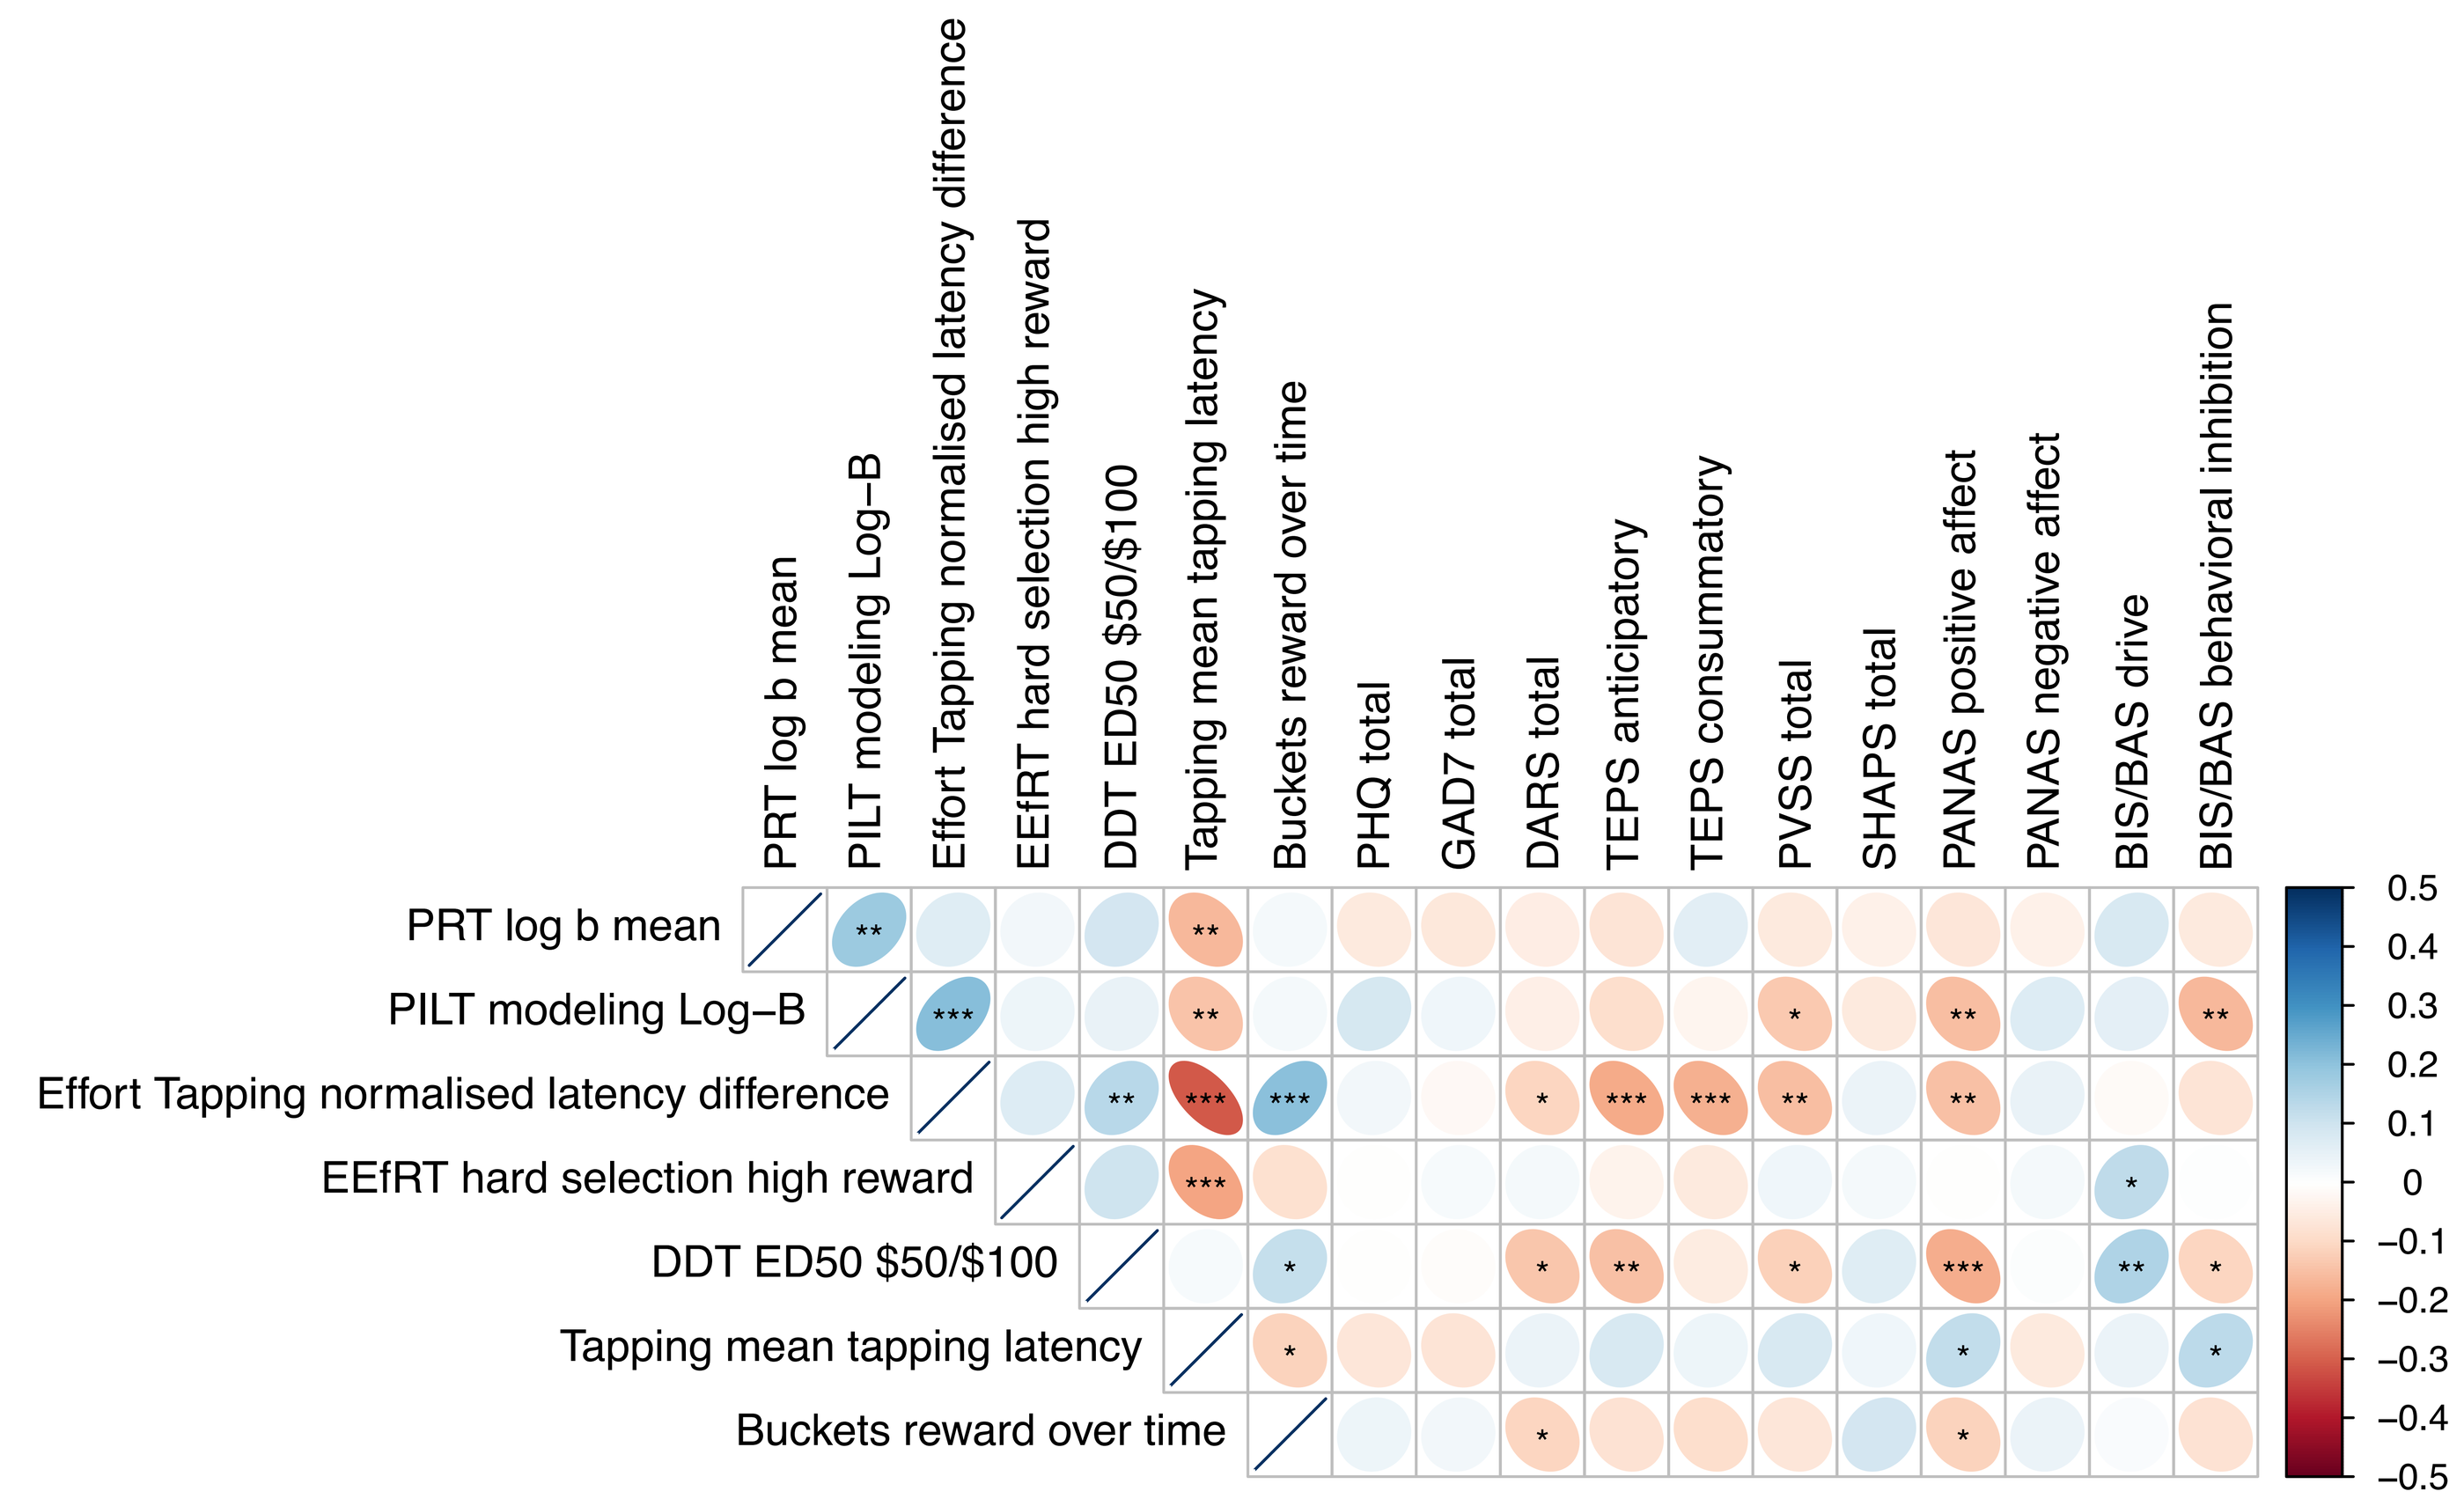
PRT log b mean
PILT modeling Log–B
Effort Tapping normalised latency difference
EEfRT hard selection high reward
DDT ED50 $50/$100
Tapping mean tapping latency
Buckets reward over time
PHQ total
GAD7 total
DARS total
TEPS anticipatory
TEPS consummatory
PVSS total
SHAPS total
PANAS positive affect
PANAS negative affect
BIS/BAS drive
BIS/BAS behavioral inhibition

Figure 4. Spearman correlations: * p < 0.05, ** p < 0.01, *** p < 0.001. Blue correlations are positive, red negative and white near zero.

8. Learning Rates and Effort Dynamics

- Figures 5 and 6 show learning across α and β modeling parameters for PILT, and effort tapping dynamics for three tertiles.

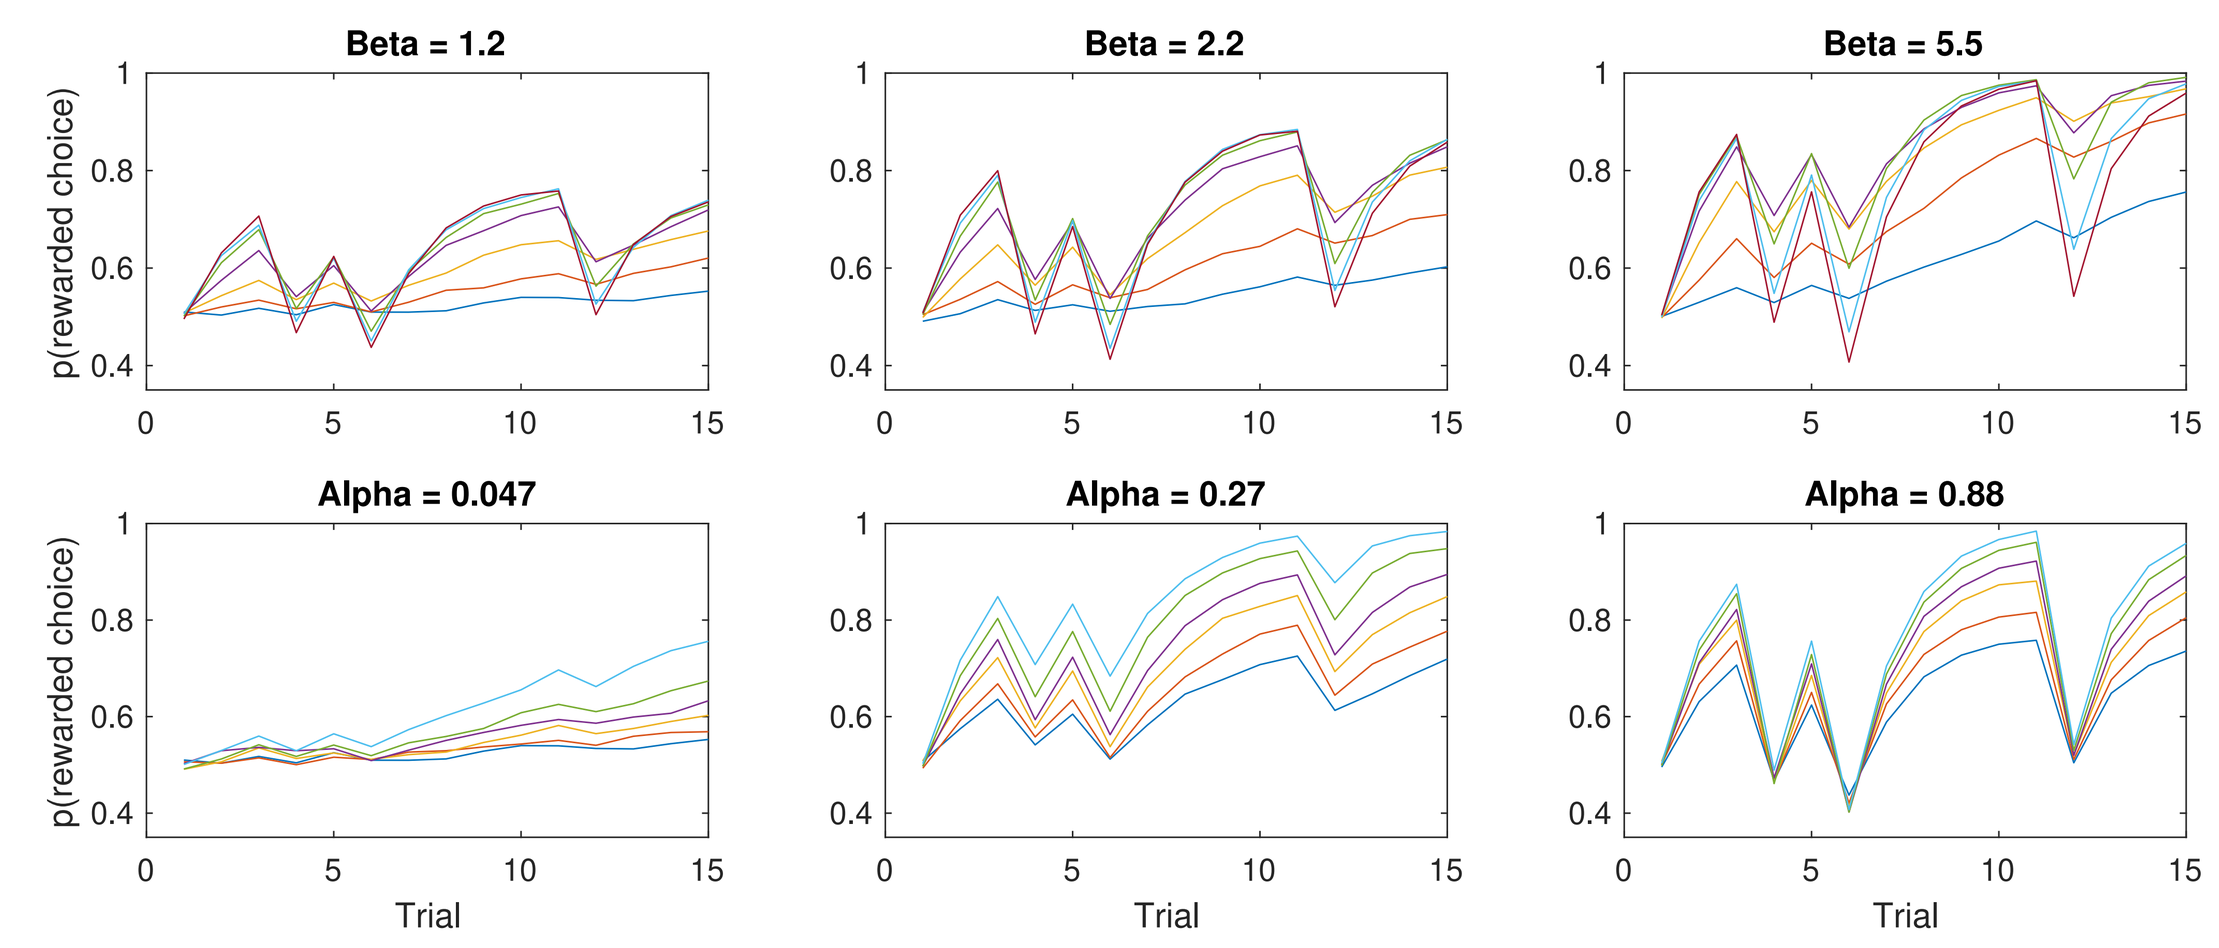
Beta = 1.2
Beta = 2.2
Beta = 5.5
Alpha = 0.047
Alpha = 0.27
Alpha = 0.88
p(rewarded choice)
Trial

Figure 5. PILT modeling: Each panel shows learning curves, i.e., the average probability of choosing the more rewarded stimulus over trials. The top row illustrates three levels of determinism (low to high), with curves for different learning rates (blue = slow, purple = fast). The bottom row presents the same data arranged by learning speed (left = slow, right = fast), with lines showing determinism levels (dark blue = low, cyan = high). Feedback was misleading on trials 3, 5, and 11, with 15 total trials.

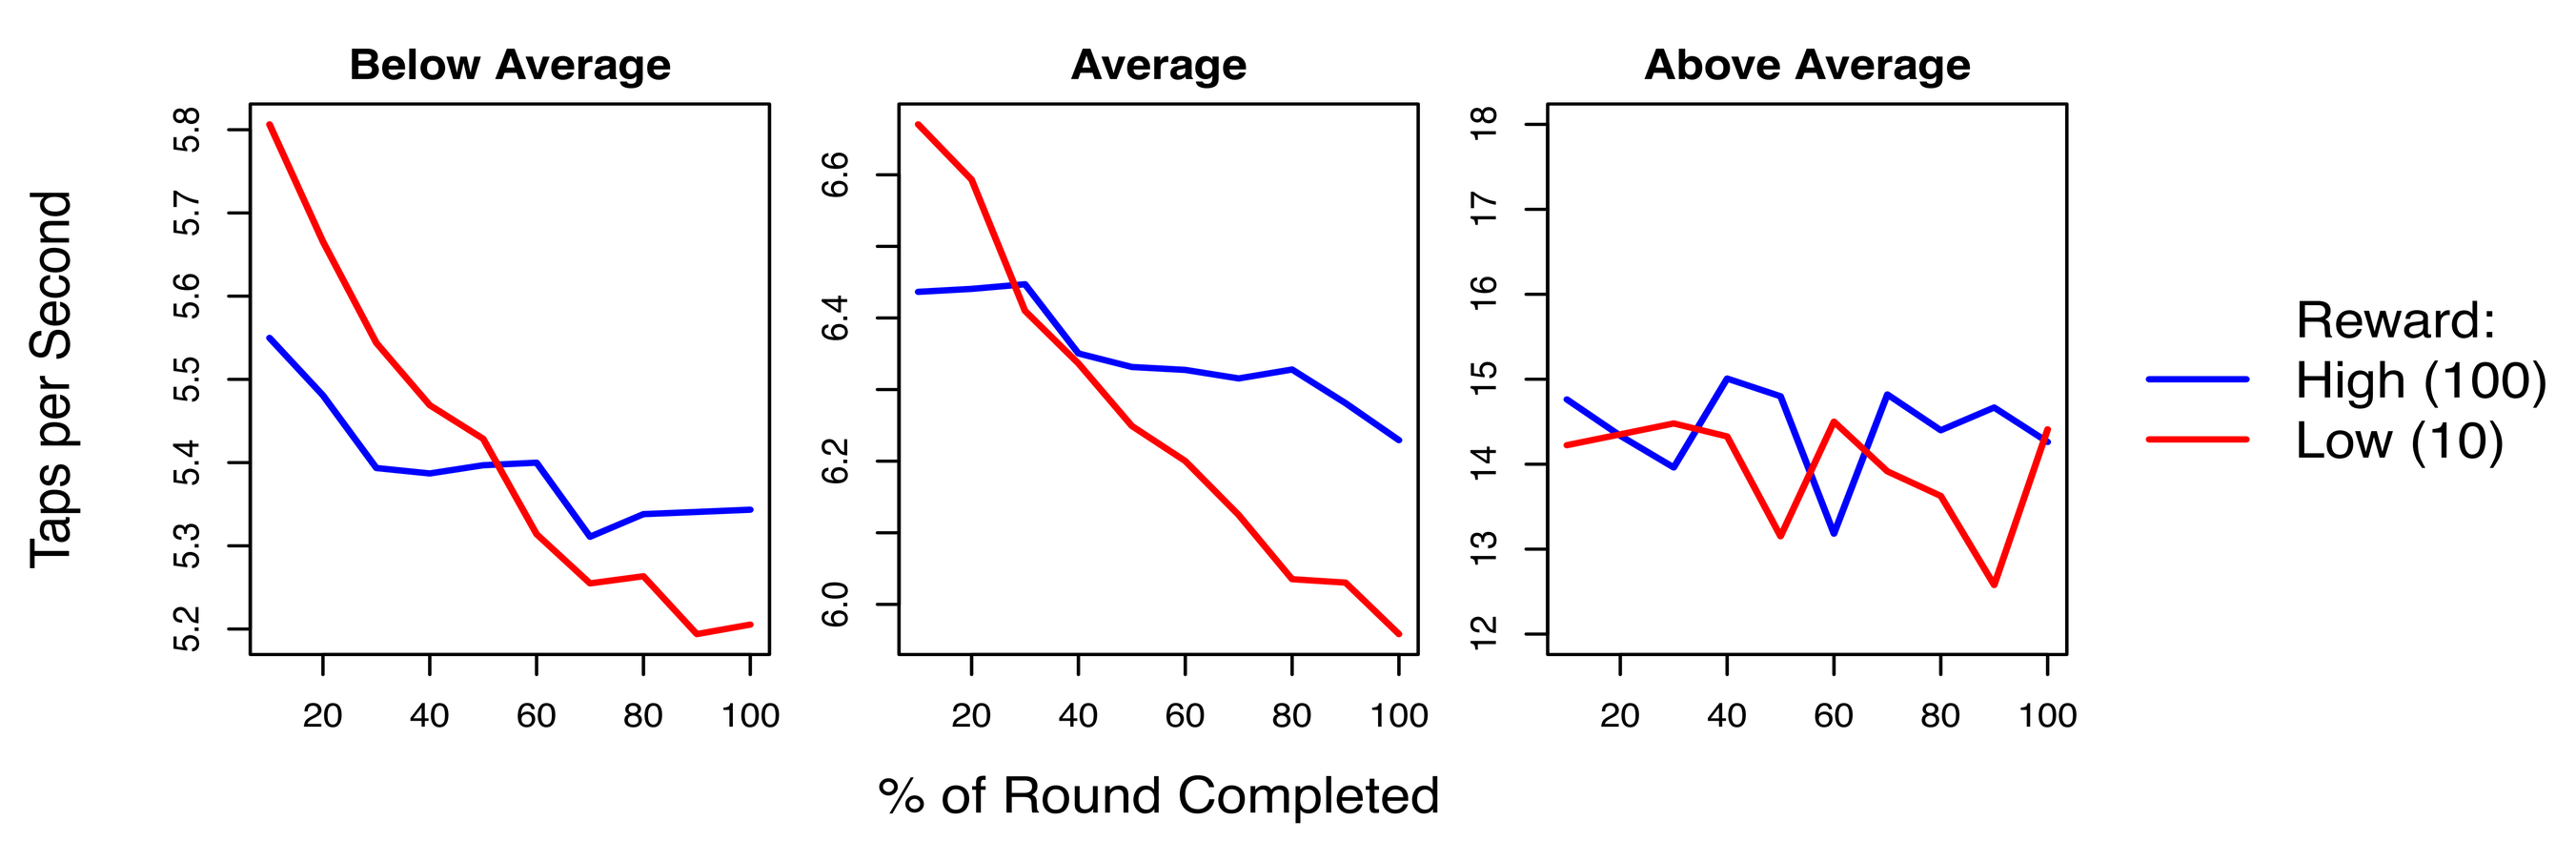
Below Average
Average
Above Average
Taps per Second
% of Round Completed
Reward:
High (100)
Low (10)

Figure 6. Effort Tapping modeling: response rate curves over the time course of round 1 for subjects with below, typical and above average tapping speed. The low tapping rate decays over time especially for slower tappers. For faster tappers the low reward tapping is slower but doesn’t decay to the same extent.

9. Conclusions

- The two new computerized cognitive tasks measuring reward learning and reward sensitivity show excellent test-retest reliability, and are shorter, better-tolerated, and display stronger evidence of construct and external validity through association with anhedonia and motivation than established reward tasks. Associations were replicated across two large studies, firstly with and then without monetary incentives for the established comparison tasks.

10. References

1. Pizzagalli DA, Jahn AL, O'Shea JP. Biol Psychiatry. 2005.
2. Treadway MT, et. al. J Abnorm Psychol. 2012.
3. Nair A, et. al. Psychological Medicine. 2023.
4. Pessiglione M, et. al. Nature. 2006.
5. Rizvi SJ, et. al. Psychiatry Res. 2015.
6. Khazanov GK, Ruscio AM, Forbes CN. Assessment. 2020.
7. Carver, CS, and White, TL. J. Pers. Soc. Psychol. 1994.
8. Zhou H, et. al. Front Psychol. 2019.
9. Watson, D., Clark, L. A., & Tellegen, A. J. Pers and Soc. Psychol. 1998.
10. Cooper, et. al. ECNP 2024.

11. Acknowledgements

- All authors from Alto Neuroscience receive salary and equity compensation from Alto Neuroscience. A.J. Savitz holds equity in J&J. QH is employed by University College London and has received fees and equity compensation from Alto Neuroscience.